32nd ECCMID
Lisbon, Portugal
23–26 April 2022
Poster P1337

Comparison between pharmacokinetic-phramcodynamic modeling results of ceftaroline against methicillin-resistant *Staphylococcus aureus* after Time Kill and Hollow Fiber experiments

Alexia Chauzy [1], Arthur Fourmy [1], Hélène Mirfendereski [1,2], William Couet [1,2] and Sandrine Marchand [1,2]

[1] INSERM U1070, Université de Poitiers - Poitiers (France)
[2] Laboratoire de Toxicologie et Pharmacocinétique, CHU de Poitiers - Poitiers (France)

Background

- Ceftaroline is a time-dependent antibiotic but its pharmacokinetic/pharmacodynamic (PK/PD) target (%T>MIC) in critically-ill patients is still debated.
- A PKPD modeling approach should help to clarify the issue.
- **The aim of this study was to evaluate whether simple Time-Kill (TK) experiments would be sufficient, or if more complex and costly Hollow-Fiber (HF) experiments would be necessary for that.**

Materials and methods

1) Static TK curves

- Methicillin-resistant *Staphylococcus aureus* strain: Baa 1026

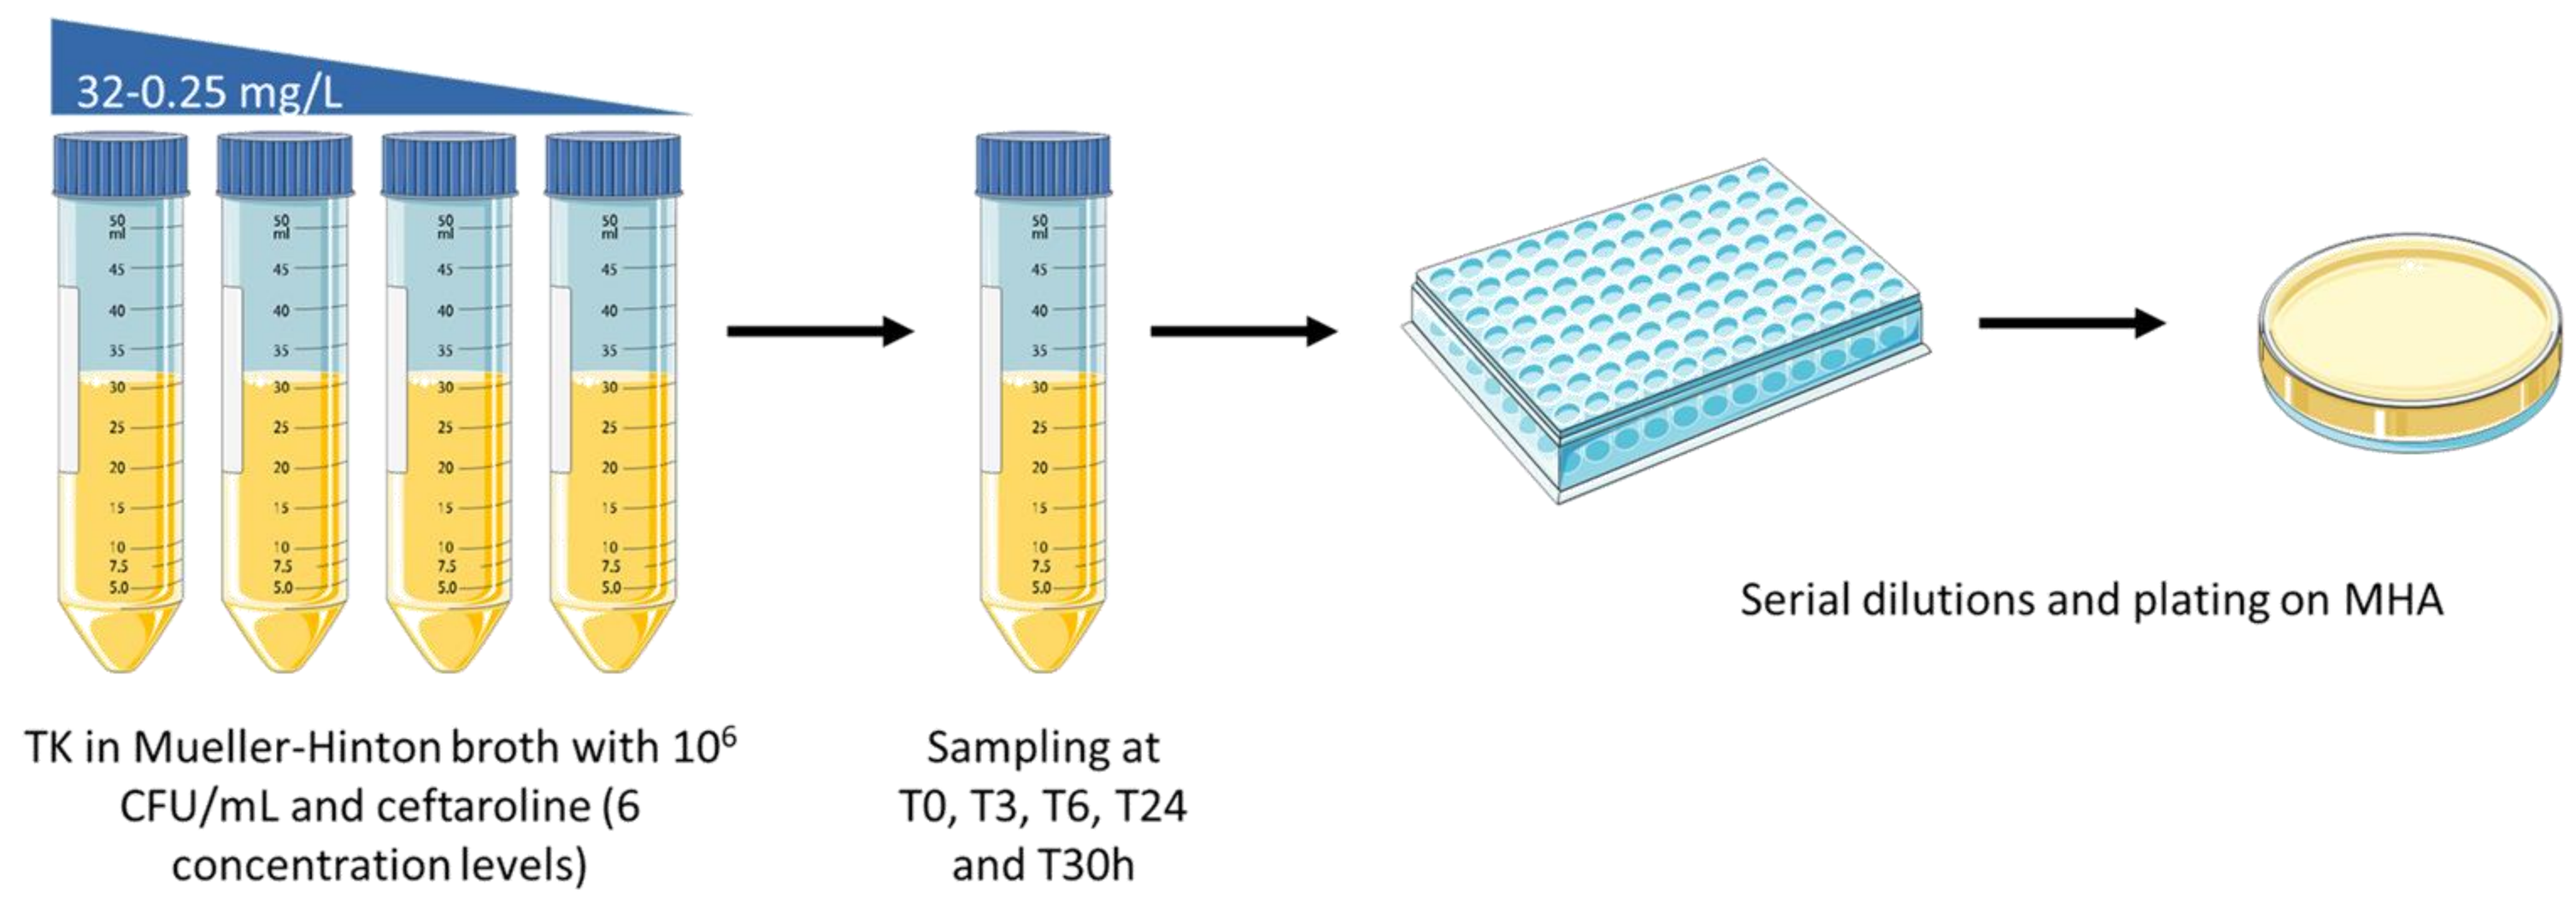
32-0.25 mg/L
Serial dilutions and plating on MHA
TK in Mueller-Hinton broth with 10^6 CFU/mL and ceftaroline (6 concentration levels)
Sampling at T0, T3, T6, T24 and T30h

2) PKPD model using NONMEM

- PD model developed from TK data
- Then combined to a population PK model of ceftaroline in ICU hyperfiltering patients *(ECCMID 2020, Abstract 02024)*
- Monte-Carlo simulations after 3 various dosing regimens corresponding respectively to 40%, 60% and 100% T>MIC

3) HF infection model (HFIM)

- PK profiles corresponding to Monte-Carlo simulations reproduced in a dynamic HFIM over 4 days
- Comparison of TK-derived simulations with HF experimental results

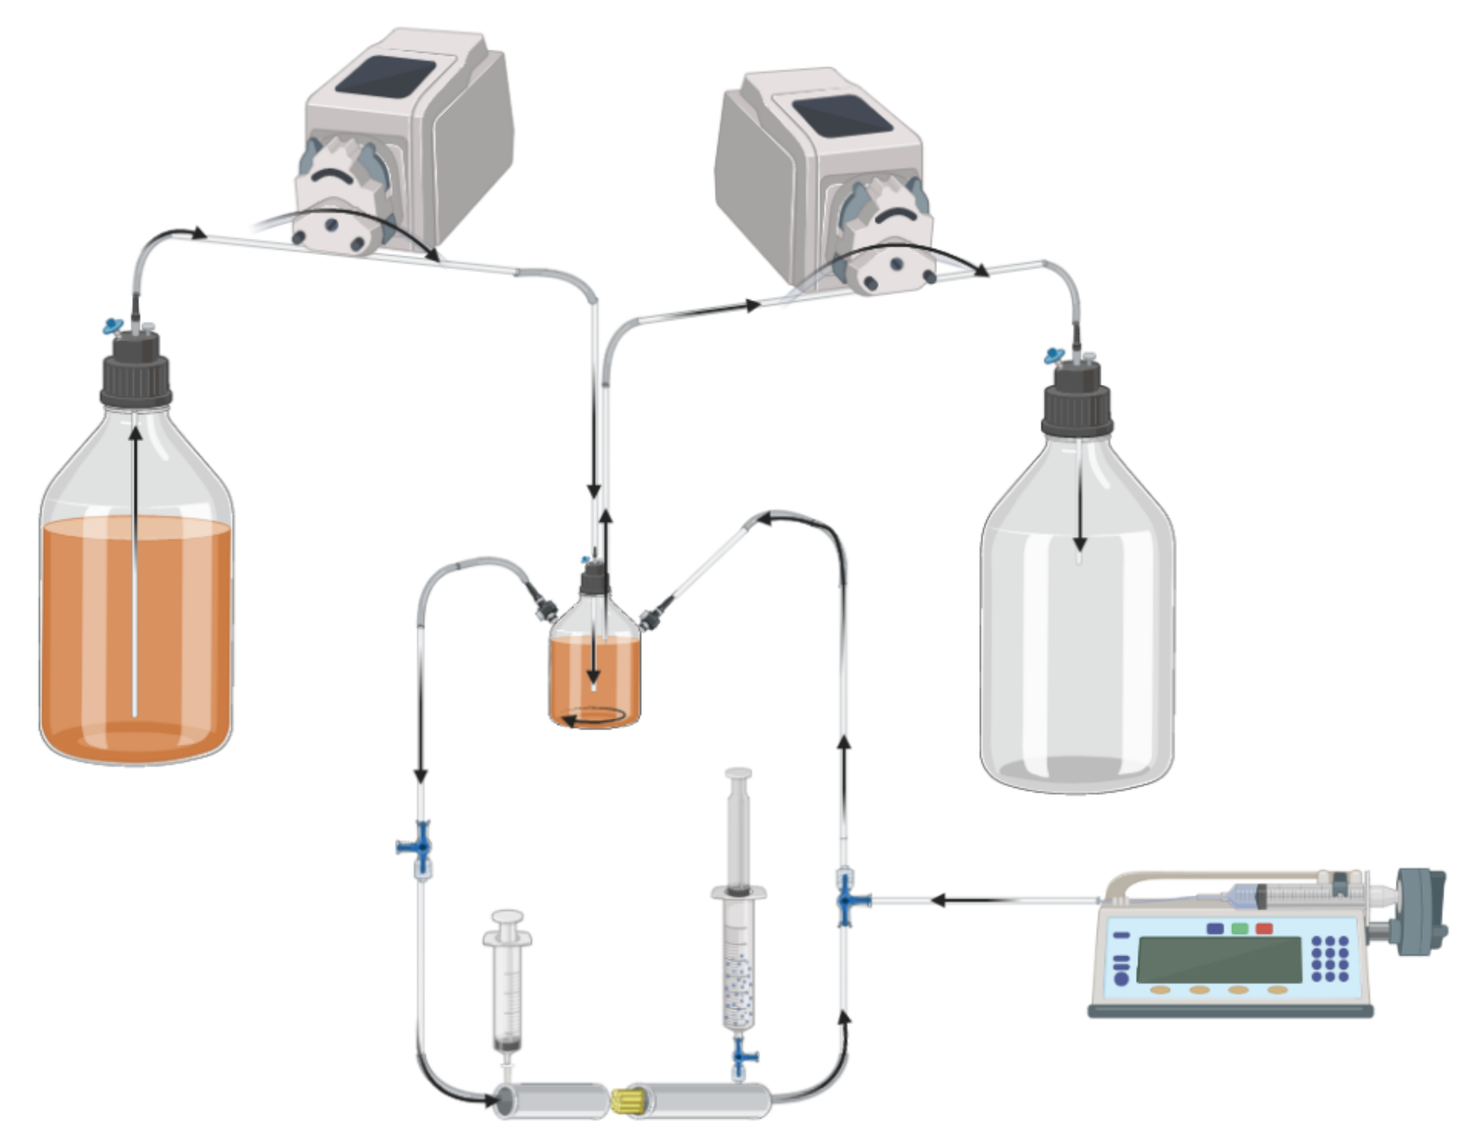

Results

1) Static TK curves

- CFU versus time data demonstrated concentration dependent decay and no regrowth (Fig.1).

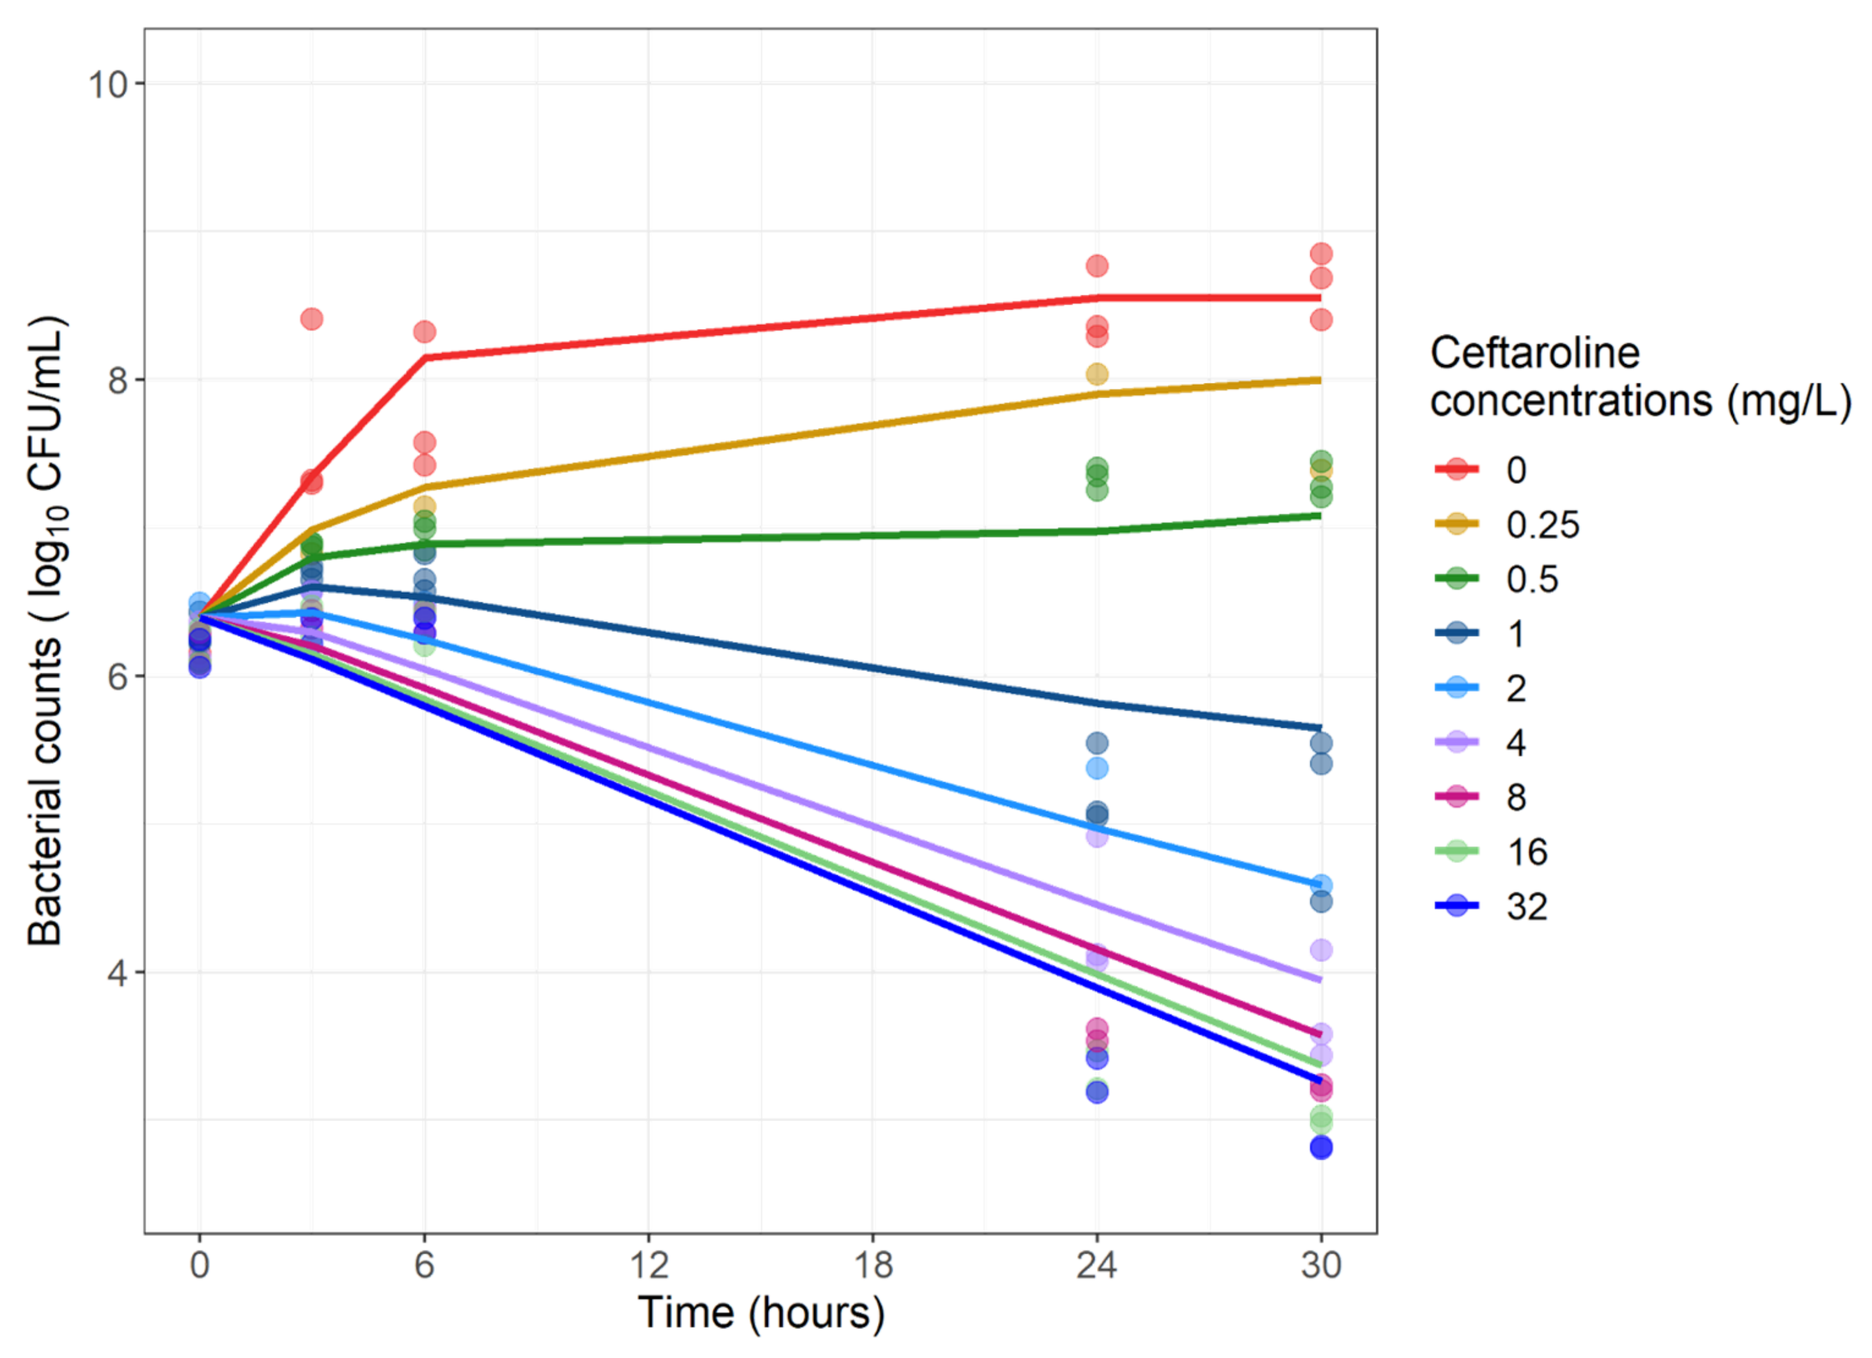
Ceftaroline concentrations (mg/L)
0
0.25
0.5
1
2
4
8
16
32
Bacterial counts ($\log_{10}$ CFU/mL)
Time (hours)

Figure 1. Static time-kill curves of ceftaroline against *S. aureus* Baa1026
Circles correspond to experimental data and lines to predictions from the model.

2) PKPD model

- TK data were best described by a one compartment model for drug-susceptible growing bacteria (Fig.2).
- Ceftaroline bactericidal effect was modeled as concentration dependent increase in the killing rate according to an Emax model.

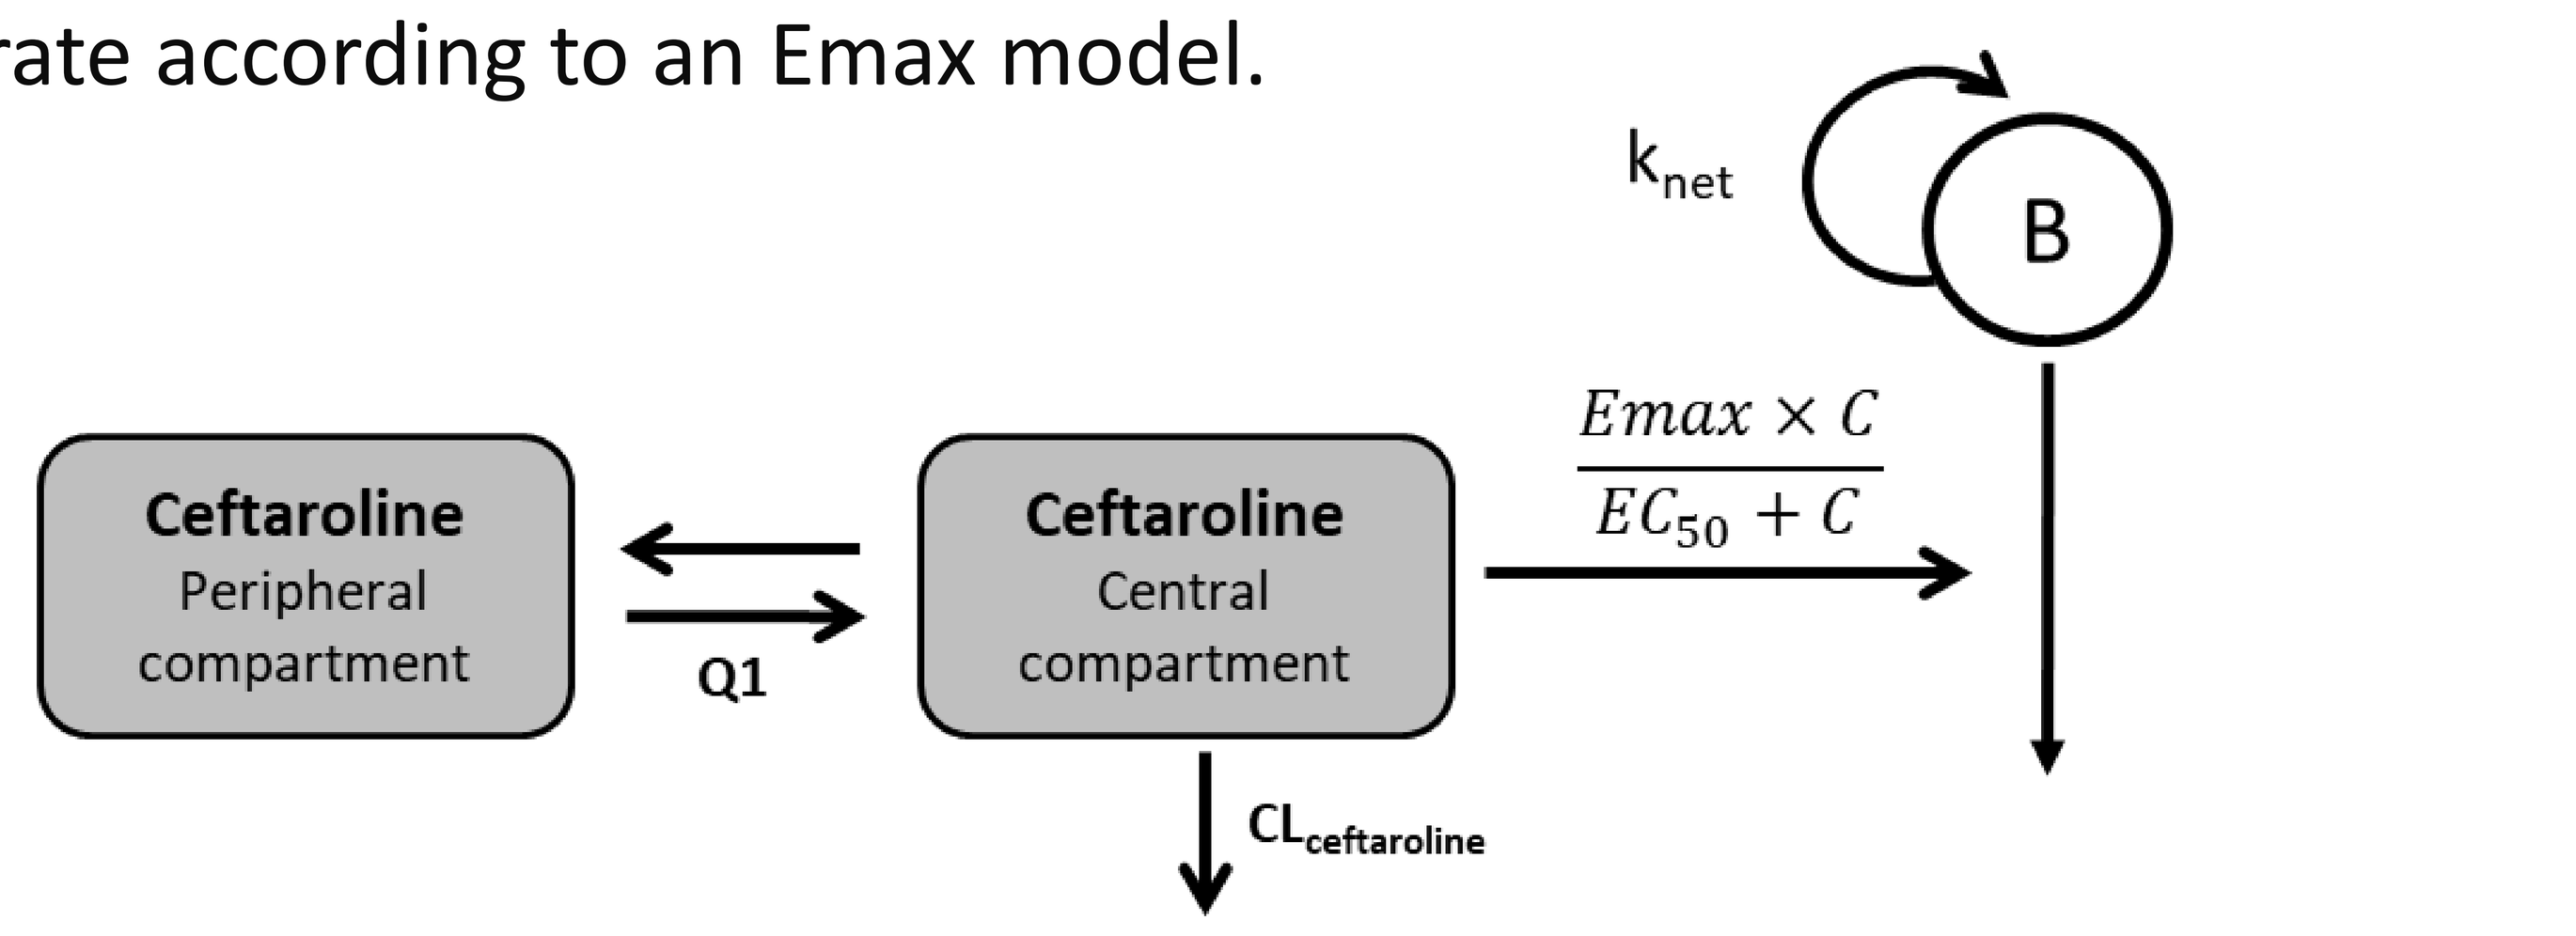
k_{net}
B
$\frac{Emax \times C}{EC_{50} + C}$
Ceftaroline Peripheral compartment
Ceftaroline Central compartment
Q1
$CL_{ceftaroline}$

Figure 2. Schematic representation of the final PKPD model used to simulate ceftaroline bactericidal effect on *S. aureus* Baa1026
Note that a two-compartments model was previously shown to adequately characterize ceftaroline PK in ICU hyperfiltering patients (ECCMID 2020, Abstract 02024) and was combined to the PD model based on TK data. With $Cl_{ceftaroline}$, ceftaroline clearance; Q, distribution clearance between central and peripheral compartments; k_{net}, apparent growth rate constant of bacteria B; C, ceftaroline concentration; E_{max}, maximum kill rate constant due to ceftaroline; EC_{50}, ceftaroline concentration that results in 50% of E_{max}

3) Comparison of TK-derived simulations with HF results

- HF experiments showed:
 - Limited transient CFU decay followed by regrowth after once a day dosing (1200mg q24h) (Fig.3B)
 - 2-$\log_{10}$ decay after approximately 24h and no regrowth after twice daily dosing (600mg q12h) (Fig.3C)
 - 2-$\log_{10}$ decay in less than 6h with no regrowth after an initial 600mg bolus dose followed by continuous infusion of 1200mg over 24h (Fig.3D)

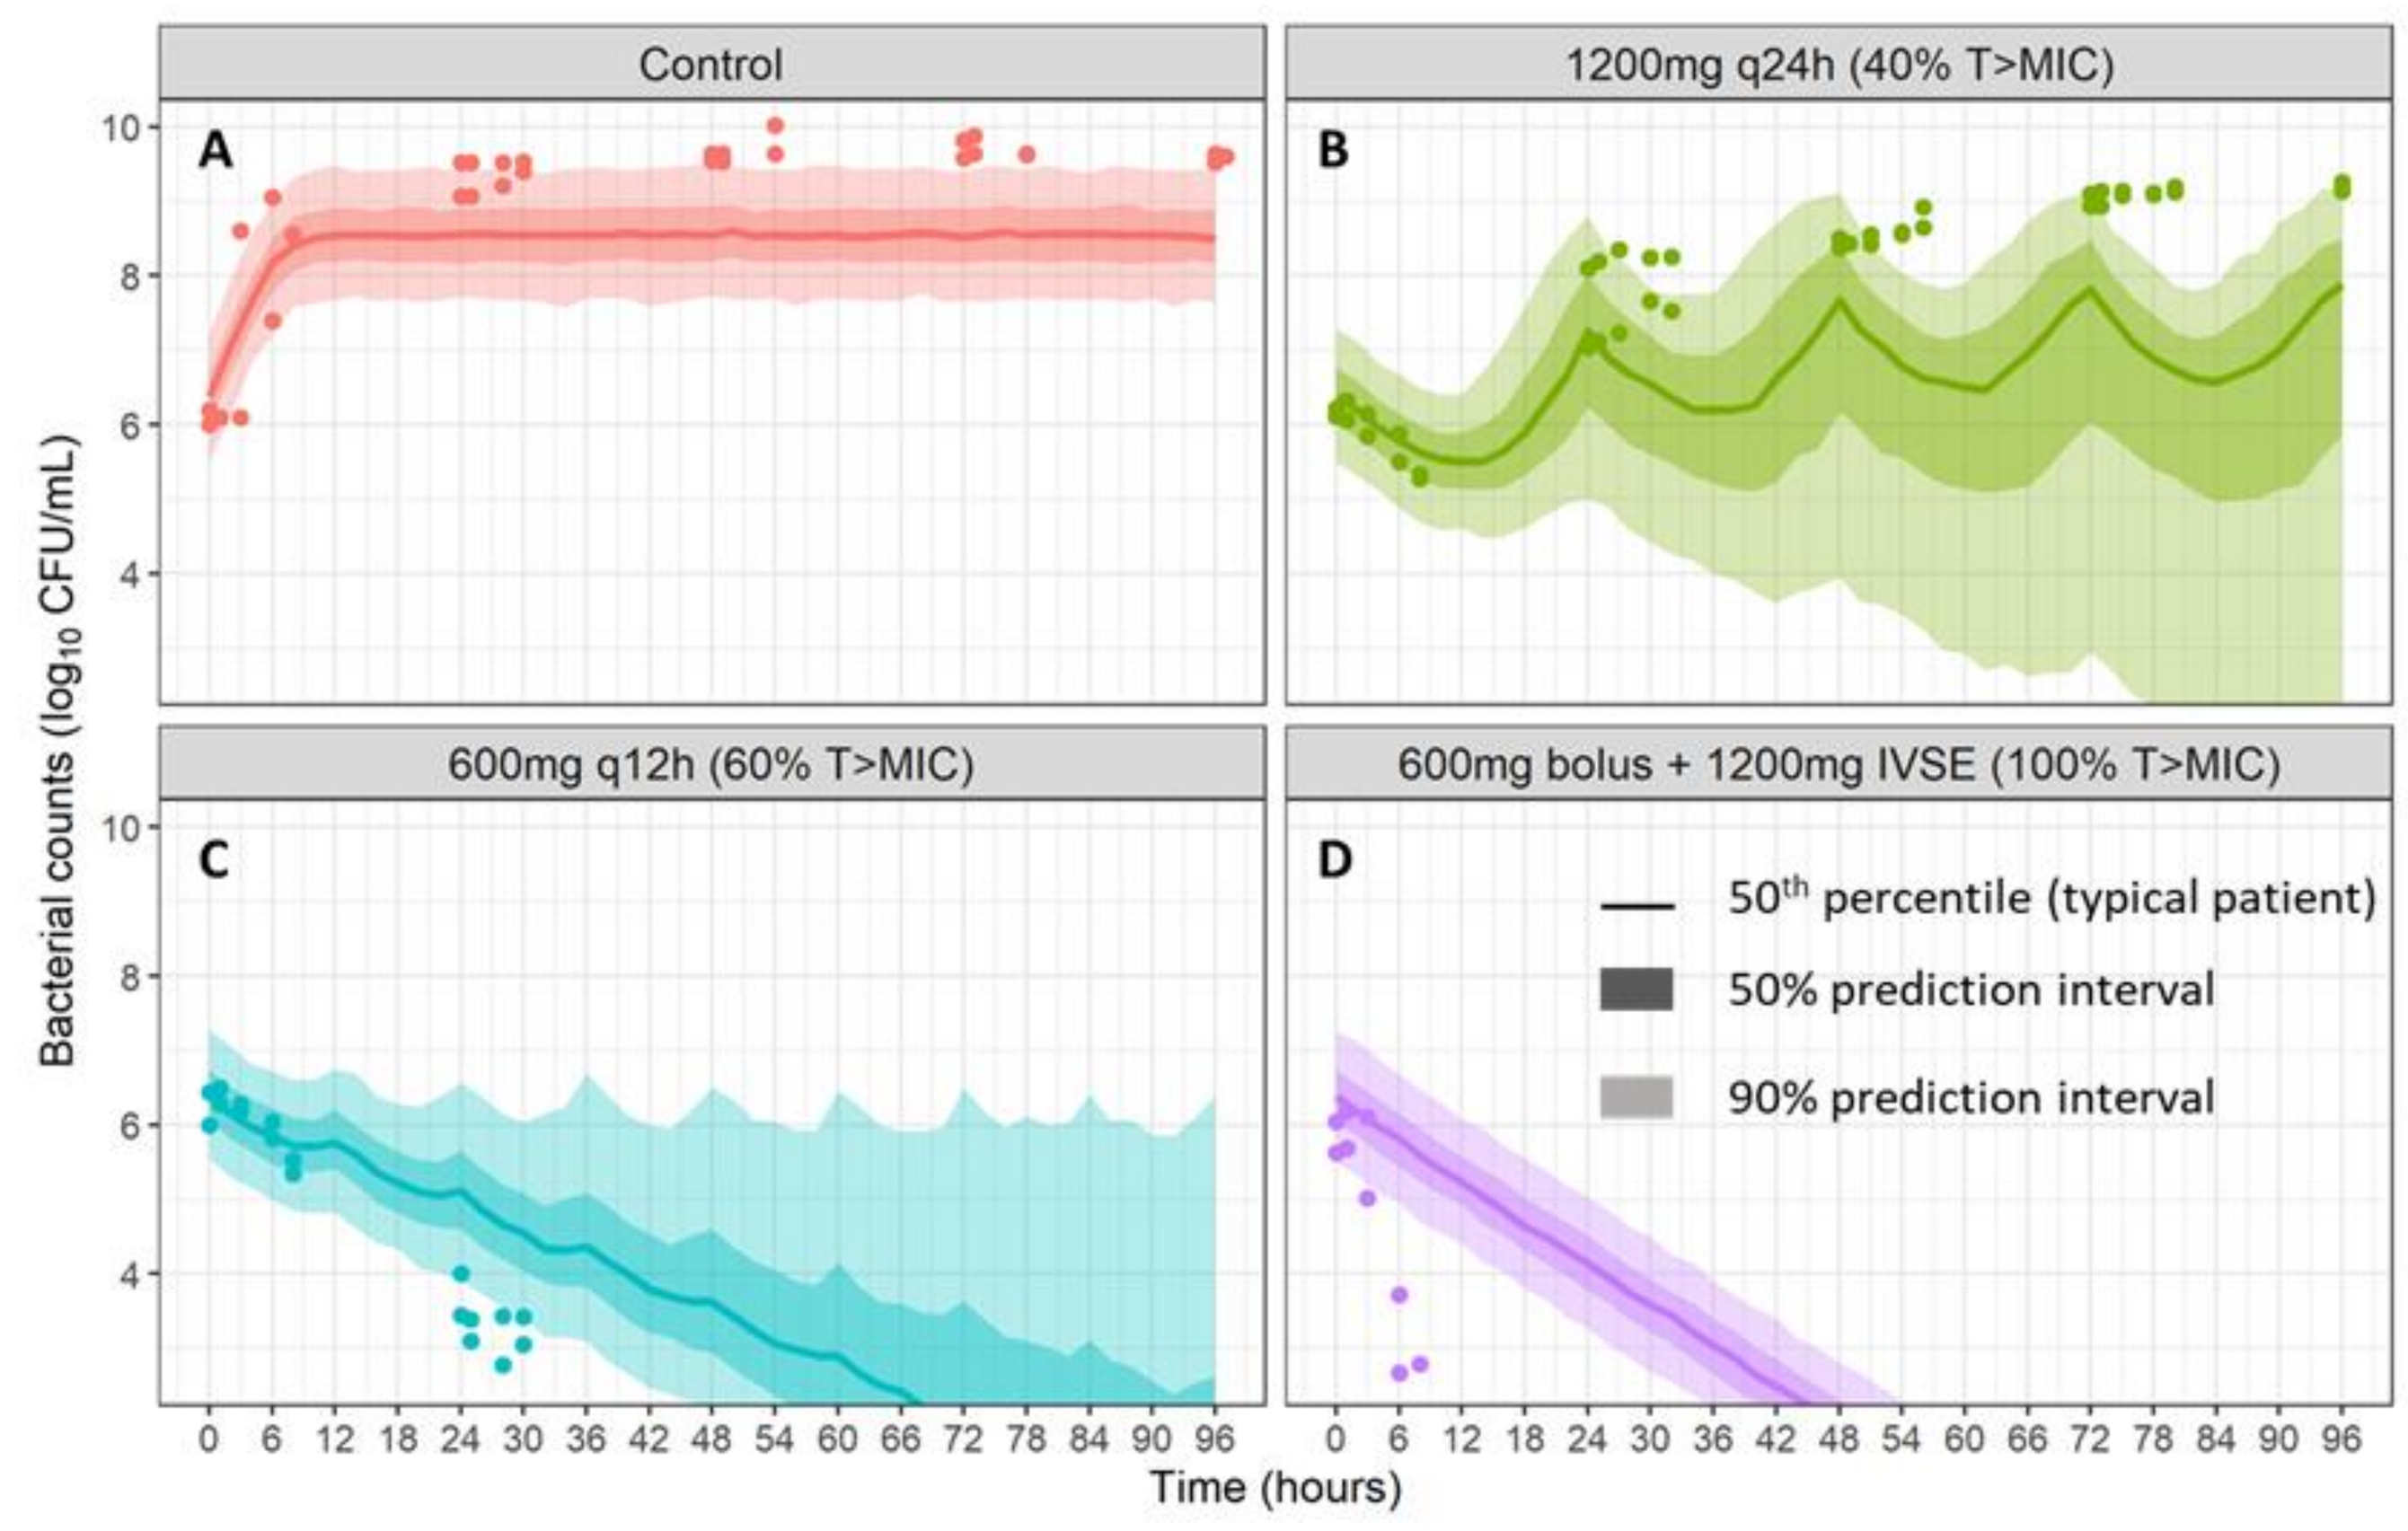
Control
1200mg q24h (40% T>MIC)
600mg q12h (60% T>MIC)
600mg bolus + 1200mg IVSE (100% T>MIC)
A
B
C
D
50^{th} percentile (typical patient)
50% prediction interval
90% prediction interval
Bacterial counts ($\log_{10}$ CFU/mL)
Time (hours)

Figure 3. Bacterial counts profiles for various dosing regimens of ceftaroline, corresponding to 0% (A), 40% (B), 60% (C) and 100% (D) T>MIC in a typical patient with creatinine clearance of 200 mL/min, against a MRSA strain.
Symbols represent observed bacterial counts in a dynamic HFIM. Straight lines and shaded areas correspond to simulated bacterial counts from the PKPD model based on TK data.

- These results were relatively, but not fully consistent with TK-derived simulations. Yet bacterial growth in the absence of antibiotic also differed between TK and HF settings (Fig.3A).
- Accordingly, a single Emax model successfully described both data sets, but parameters values derived from TK and HF experiments were somewhat different (growth rate constant and maximum kill rate constant 65% higher with HF than with TK).

Conclusion

TK experiments were able to predict the relative efficacy of the various ceftaroline dosing regimen strategies observed during HF experiments, although HF observations did not match perfectly with TK simulations.